ALRN-6924, a Dual Inhibitor of MDMX and MDM2, Transiently Induces Cell Cycle Arrest in Bone Marrow and Prevents Toxicity in Mouse Models of Acute Radiation Injury

D. Allen Annis, David Sutton, Manuel Aivado, Vojislav Vukovic Aileron Therapeutics Inc., Watertown, MA

Background

Myelosuppression is a common sequela of acute radiation injury due to sensitivity of proliferating bone marrow cells to ionizing radiation. ALRN-6924 is a clinical-stage, first-in-class, stabilized cell-permeating alpha-helical peptide drug that disrupts the interaction of the p53 tumor suppressor protein with its endogenous inhibitors, MDMX and MDM2, to induce transient, dose-dependent cell cycle arrest in p53-wild-type tissues. ALRN-6924 is being evaluated in clinical trials as a selective chemoprotection agent for patients with p53-mutant cancers to protect healthy normal cells from chemotherapy while not protecting p53-mutant cancer cells. We tested whether ALRN-6924 may similarly protect against radiation-induced toxicity in mouse models of acute radiation injury.

Materials and Methods

Activation of p21 (CDKN1A), a cell cycle regulator under transcriptional control of p53, was measured in formalin-fixed mouse bone marrow by immunohistochemistry analysis (IHC). Proliferation and apoptosis in bone marrow were measured by IHC of Ki67 and cleaved PARP, respectively. Cell cycle arrest was measured in the bone marrow of ALRN-6924-treated C57BL/6 mice by flow cytometry using EdU incorporation. Serum levels of macrophage inhibitory cytokine-1 (MIC-1), a biomarker of p53 activation, were measured by ELISA. As a model of radiation-induced toxicity, C57BL/6 mice (n=7/group) were treated with one or more intravenous 2.4 mg/kg doses of ALRN-6924 at 24, 16, 8, or 1 hour (or combinations thereof) or placebo prior to a 15 Gy shielded-body radiation dose and then monitored for body weight (BW) changes.

Results

ALRN-6924 induced cell cycle arrest in mouse bone marrow with a maximum effect at 8 hrs post-dose. Repeated doses of ALRN-6924 every 8 hrs elevated p21 levels in bone marrow that correlated with reduced Ki67 positivity and increased serum MIC-1 levels. Treatment-dependent changes in cPARP expression in bone marrow were evident, but minimal in magnitude. In a nonlethal radiation exposure model, ALRN-6924 yielded significant protection from radiation-induced BW loss in a schedule-dependent manner. Placebo-treated mice showed 10% to 15% BW loss five days after irradiation, while mice receiving one or more ALRN-6924 doses 8 hrs prior to irradiation had an average of 4% BW loss (p=0.008, two-sided t test).

Figure 1. Structure and Key Design Properties of ALRN-6924

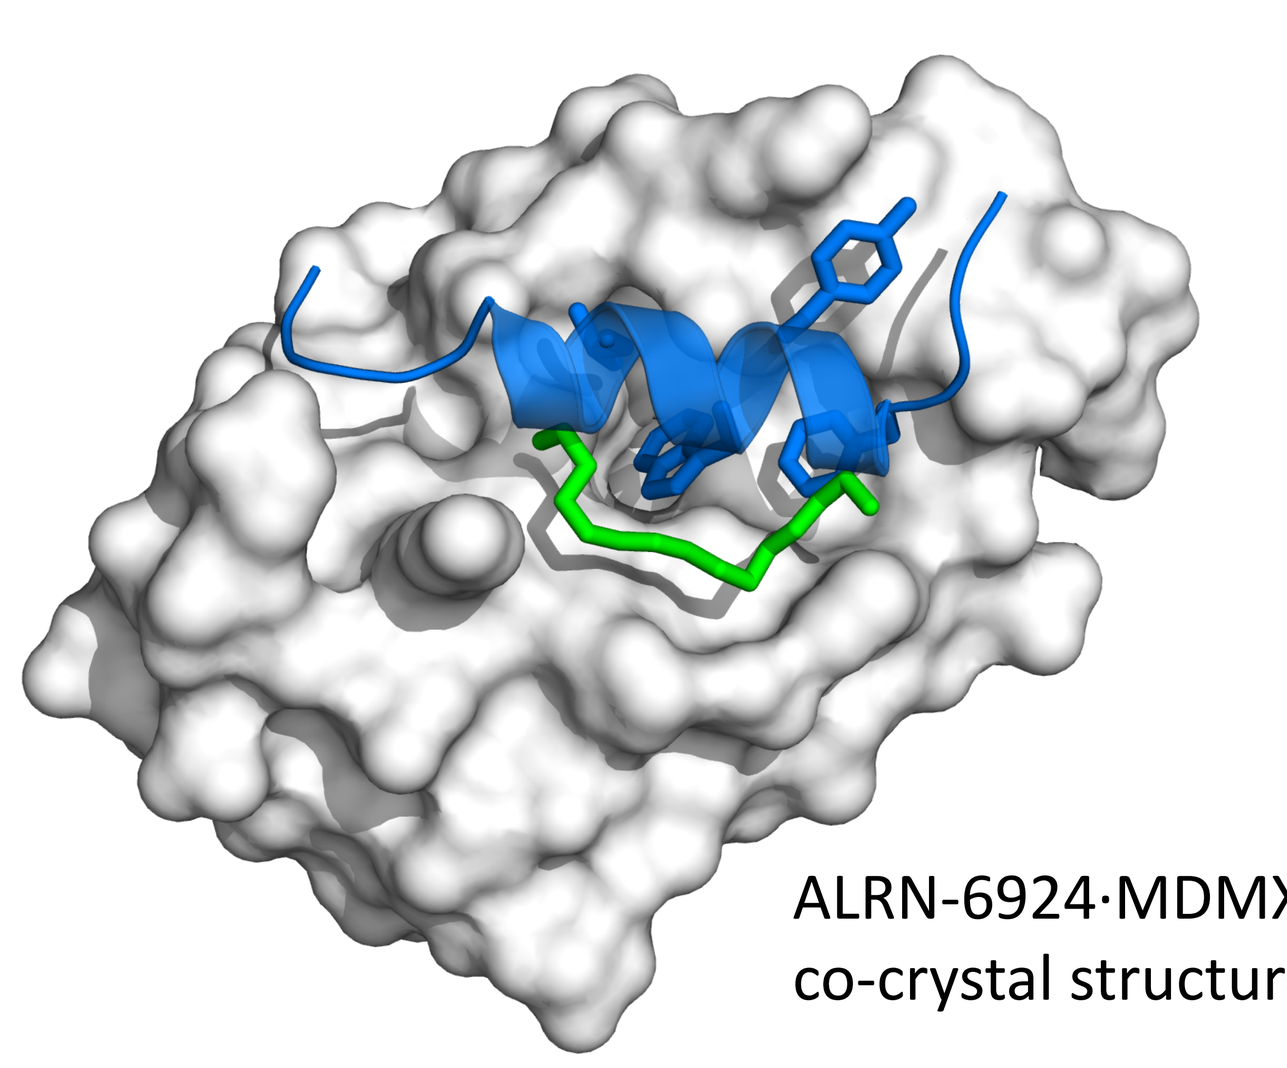

ALRN-6924·MDMX co-crystal structure

Structure is based on the N-terminal α-helical domain of p53, with a hydrocarbon staple and other chemical modifications to ensure:

- I Protection from proteolytic cleavage
- II Permeation of cell membranes and cell entry
- III High affinity binding to its targets
- IV Preclinical and clinical on-target, on-mechanism effects

Figure 2. ALRN-6924 Induces Cell Cycle Arrest in Mouse Bone Marrow that Correlates with Serum Biomarker MIC-1

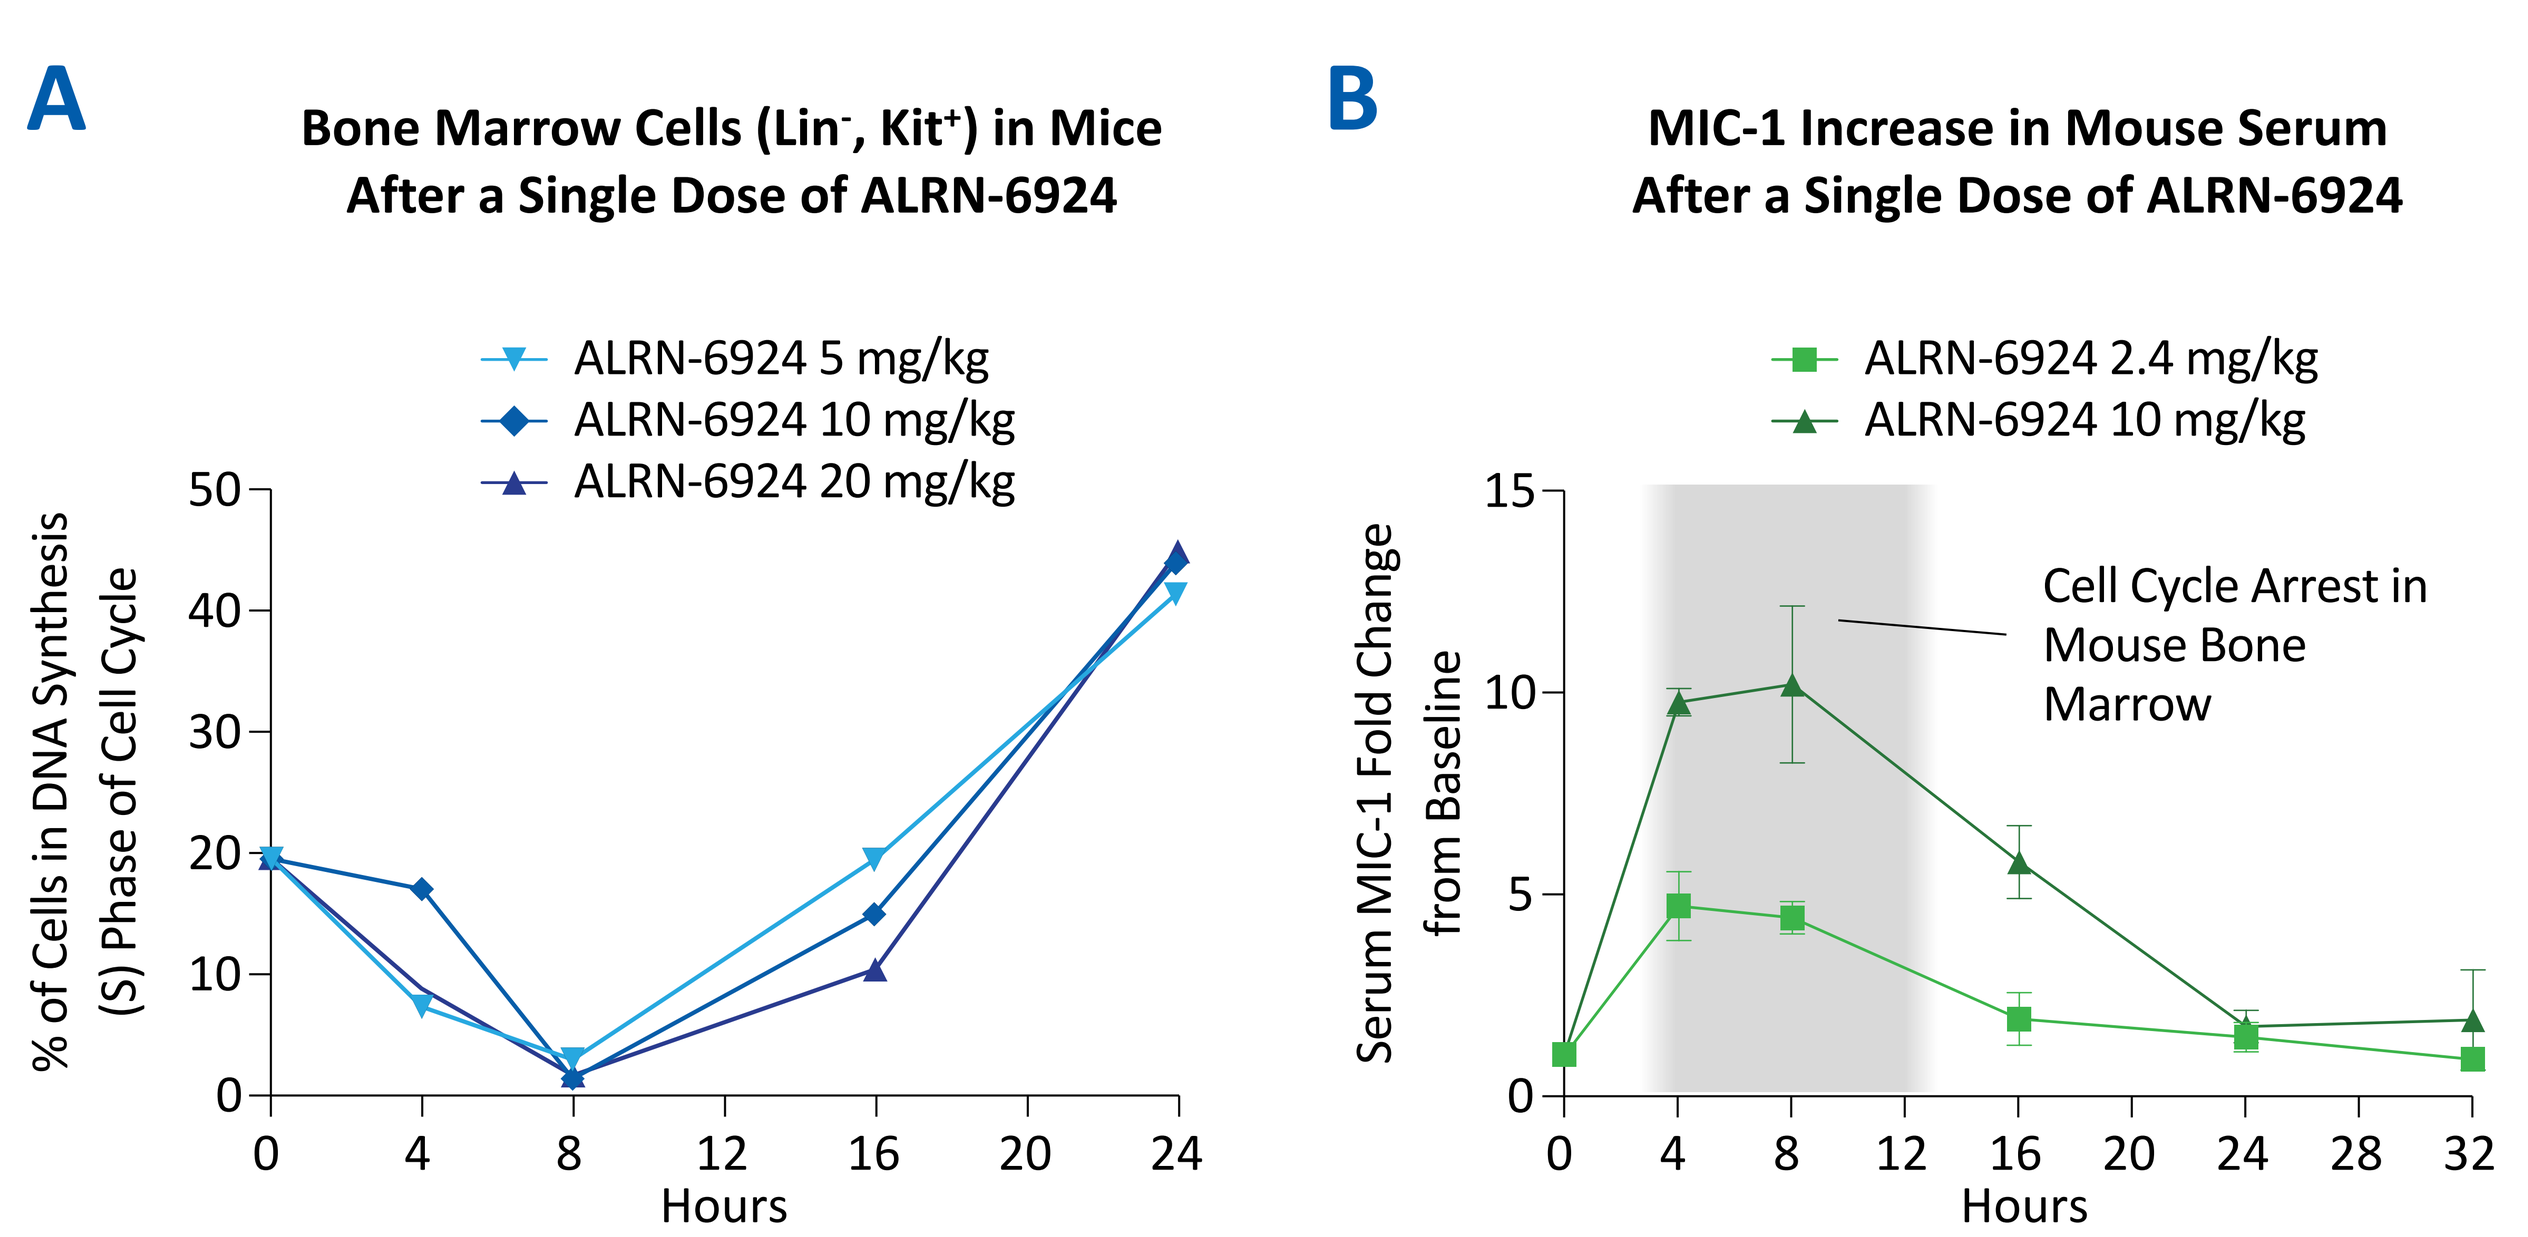
A
Bone Marrow Cells (Lin-, Kit+) in Mice After a Single Dose of ALRN-6924
ALRN-6924 5 mg/kg
ALRN-6924 10 mg/kg
ALRN-6924 20 mg/kg
% of Cells in DNA Synthesis (S) Phase of Cell Cycle
Hours
B
MIC-1 Increase in Mouse Serum After a Single Dose of ALRN-6924
ALRN-6924 2.4 mg/kg
ALRN-6924 10 mg/kg
Cell Cycle Arrest in Mouse Bone Marrow
Serum MIC-1 Fold Change from Baseline
Hours

A) Cell cycle arrest in the bone marrow of ALRN-6924-treated C57BL/6 mice reached a maximum at 8 hrs after treatment in lineage-negative, c-Kit positive hematopoietic stem and progenitor cells.

B) Levels of serum MIC-1, a biomarker of p53 activation, peak between 4 and 8 hours after treatment with ALRN-6924 *in vivo*, which correlates with cell cycle arrest in bone marrow.

Figure 3. Repeated Doses of ALRN-6924 Every 8 Hrs Yield Sustained MIC-1 Elevation

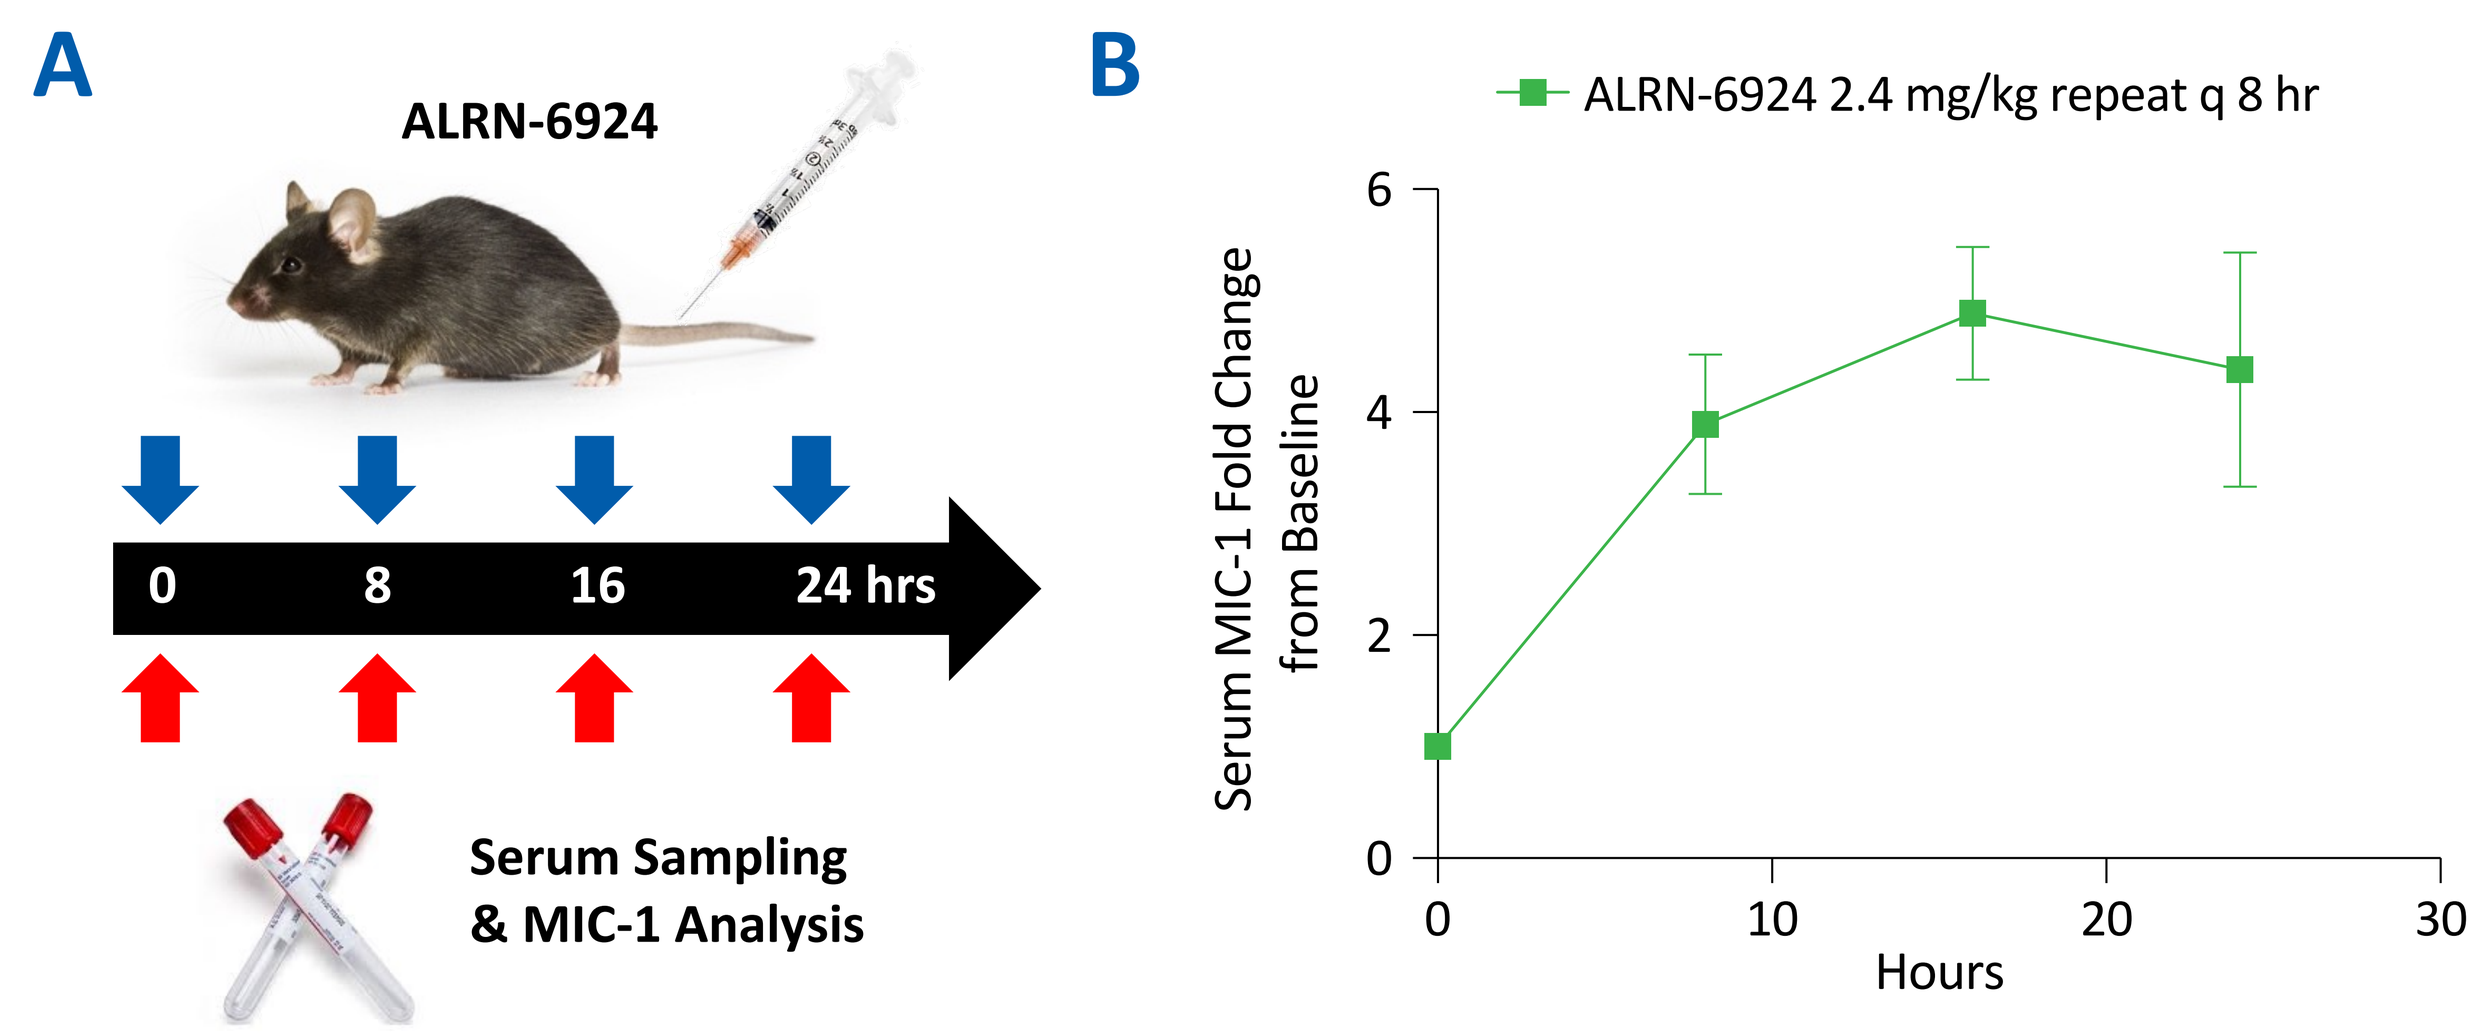
A
ALRN-6924
0
8
16
24 hrs
Serum Sampling & MIC-1 Analysis
B
ALRN-6924 2.4 mg/kg repeat q 8 hr
Serum MIC-1 Fold Change from Baseline
Hours

A) C57BL/6 mice (n=20) were administered 2.4 mg/kg ALRN-6924 every 8 hrs with n=5 sampled per time point for serum MIC-1 analysis.

B) Murine MIC-1 levels demonstrated sustained elevation following repeated dosing every 8 hrs.

Figure 4. Multiple ALRN-6924 Dose Regimens were Evaluated for Bone Marrow and GI Tract Cell Cycle Arrest, Cell Proliferation, and Apoptosis Biomarkers

Dosing Relative to Sampling

Group	Treatment	-24	-16	-8	-1	0 hr
1	Vehicle					
2	ALRN-6924 2.4mg/kg					
3	ALRN-6924 2.4mg/kg					
4	ALRN-6924 2.4mg/kg					
5	ALRN-6924 2.4mg/kg					
6	ALRN-6924 2.4mg/kg					
7	ALRN-6924 2.4mg/kg					

C57BL/6 mice (n=3/group) were treated with one or more doses/schedules of ALRN-6924 prior to collection of femur and jejunum for IHC analysis of cell proliferation (Ki67), p53-mediated cell cycle arrest (p21), and apoptosis (cPARP) biomarkers. Pathology analysis was conducted in a blinded fashion.

Figure 5. Bone Marrow Staining for Cell Proliferation Marker Ki67 was Lowest for | -8 hr | and | -16, -8 hr | Schedules

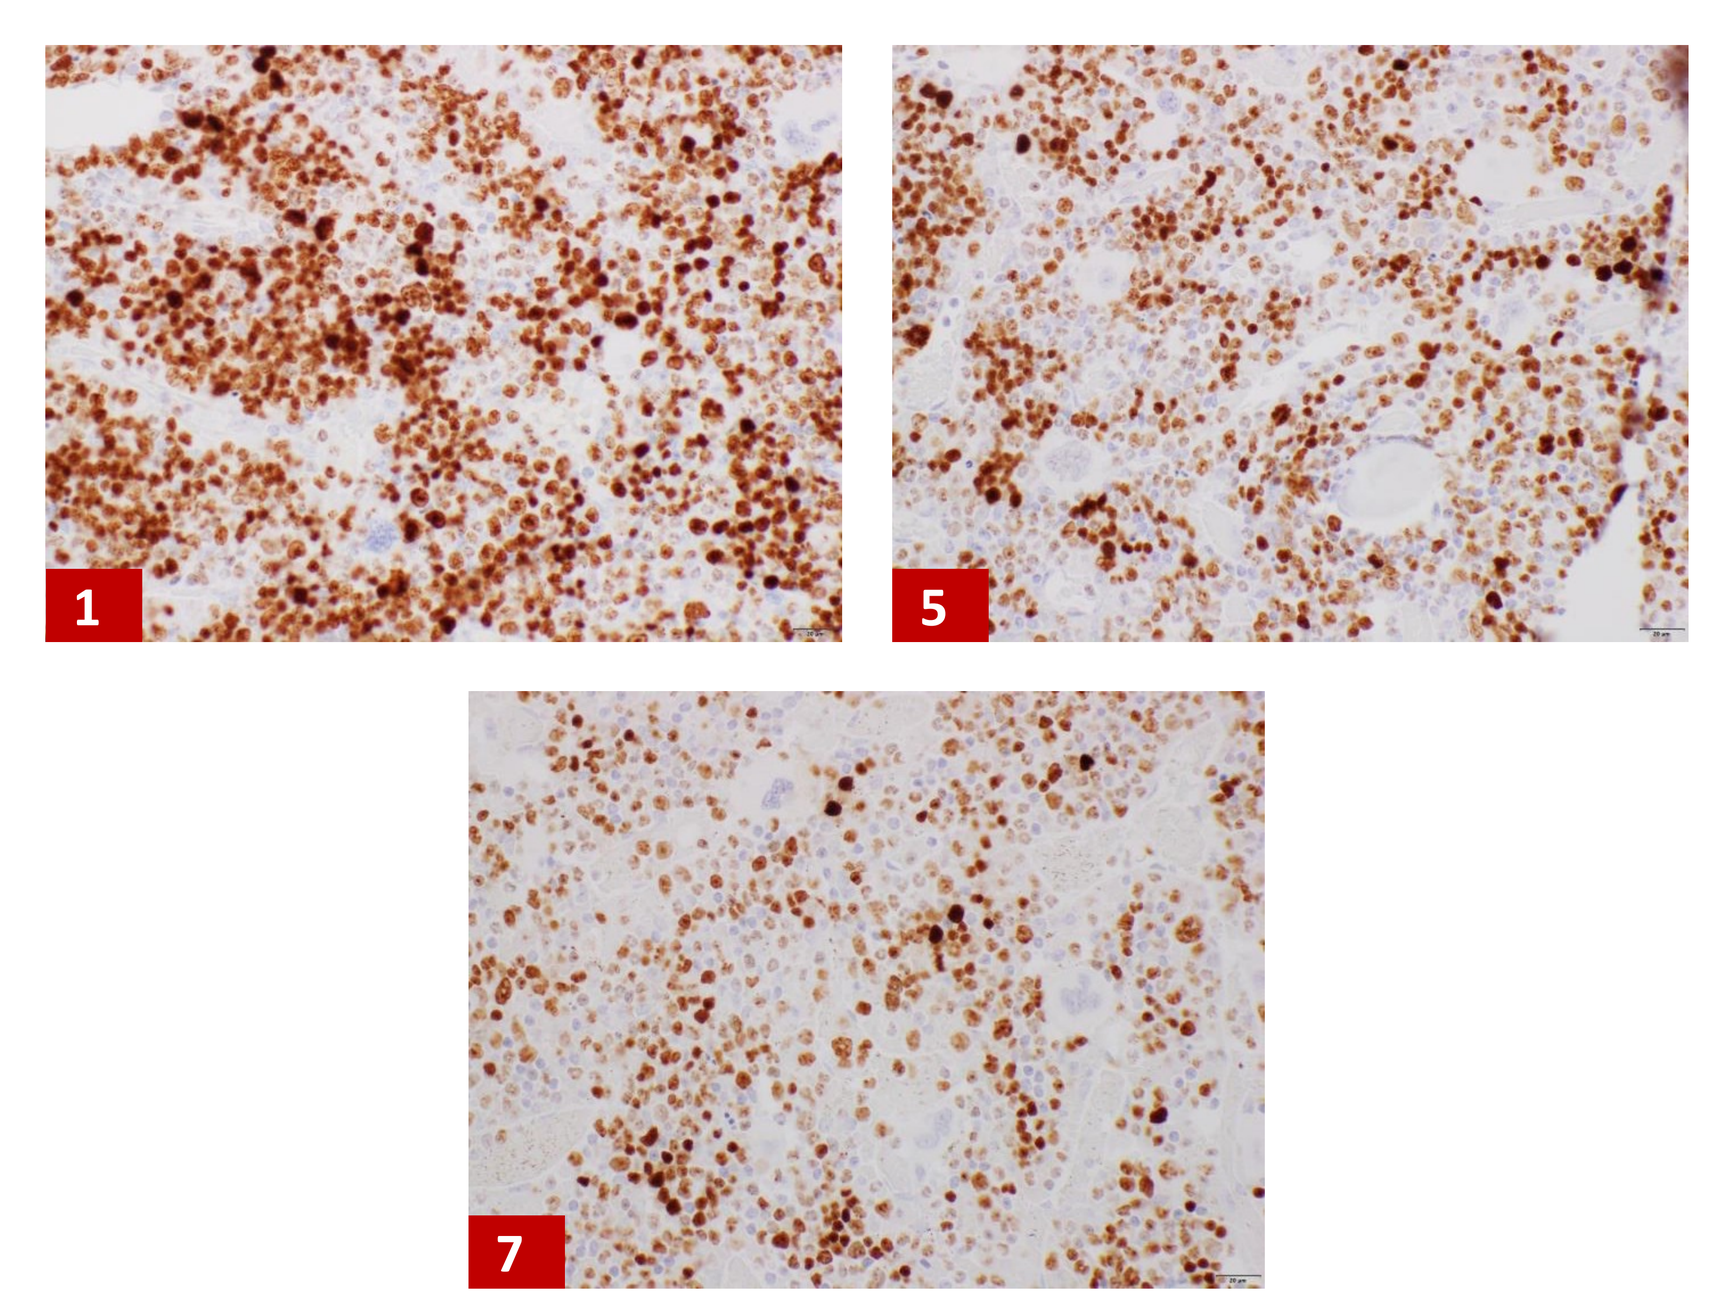
1
5
7

Dosing Relative to Sampling

Group	Treatment	-24	-16	-8	-1	0 hr
1	Vehicle					
2	ALRN-6924 2.4mg/kg					
3	ALRN-6924 2.4mg/kg					
4	ALRN-6924 2.4mg/kg					
5	ALRN-6924 2.4mg/kg					
6	ALRN-6924 2.4mg/kg					
7	ALRN-6924 2.4mg/kg					

Figure 6. Bone Marrow Staining for Cell Cycle Arrest Marker p21 was Highest for | -8 hr |, | -24, -8 hr |, | -16, -8 hr | Schedules

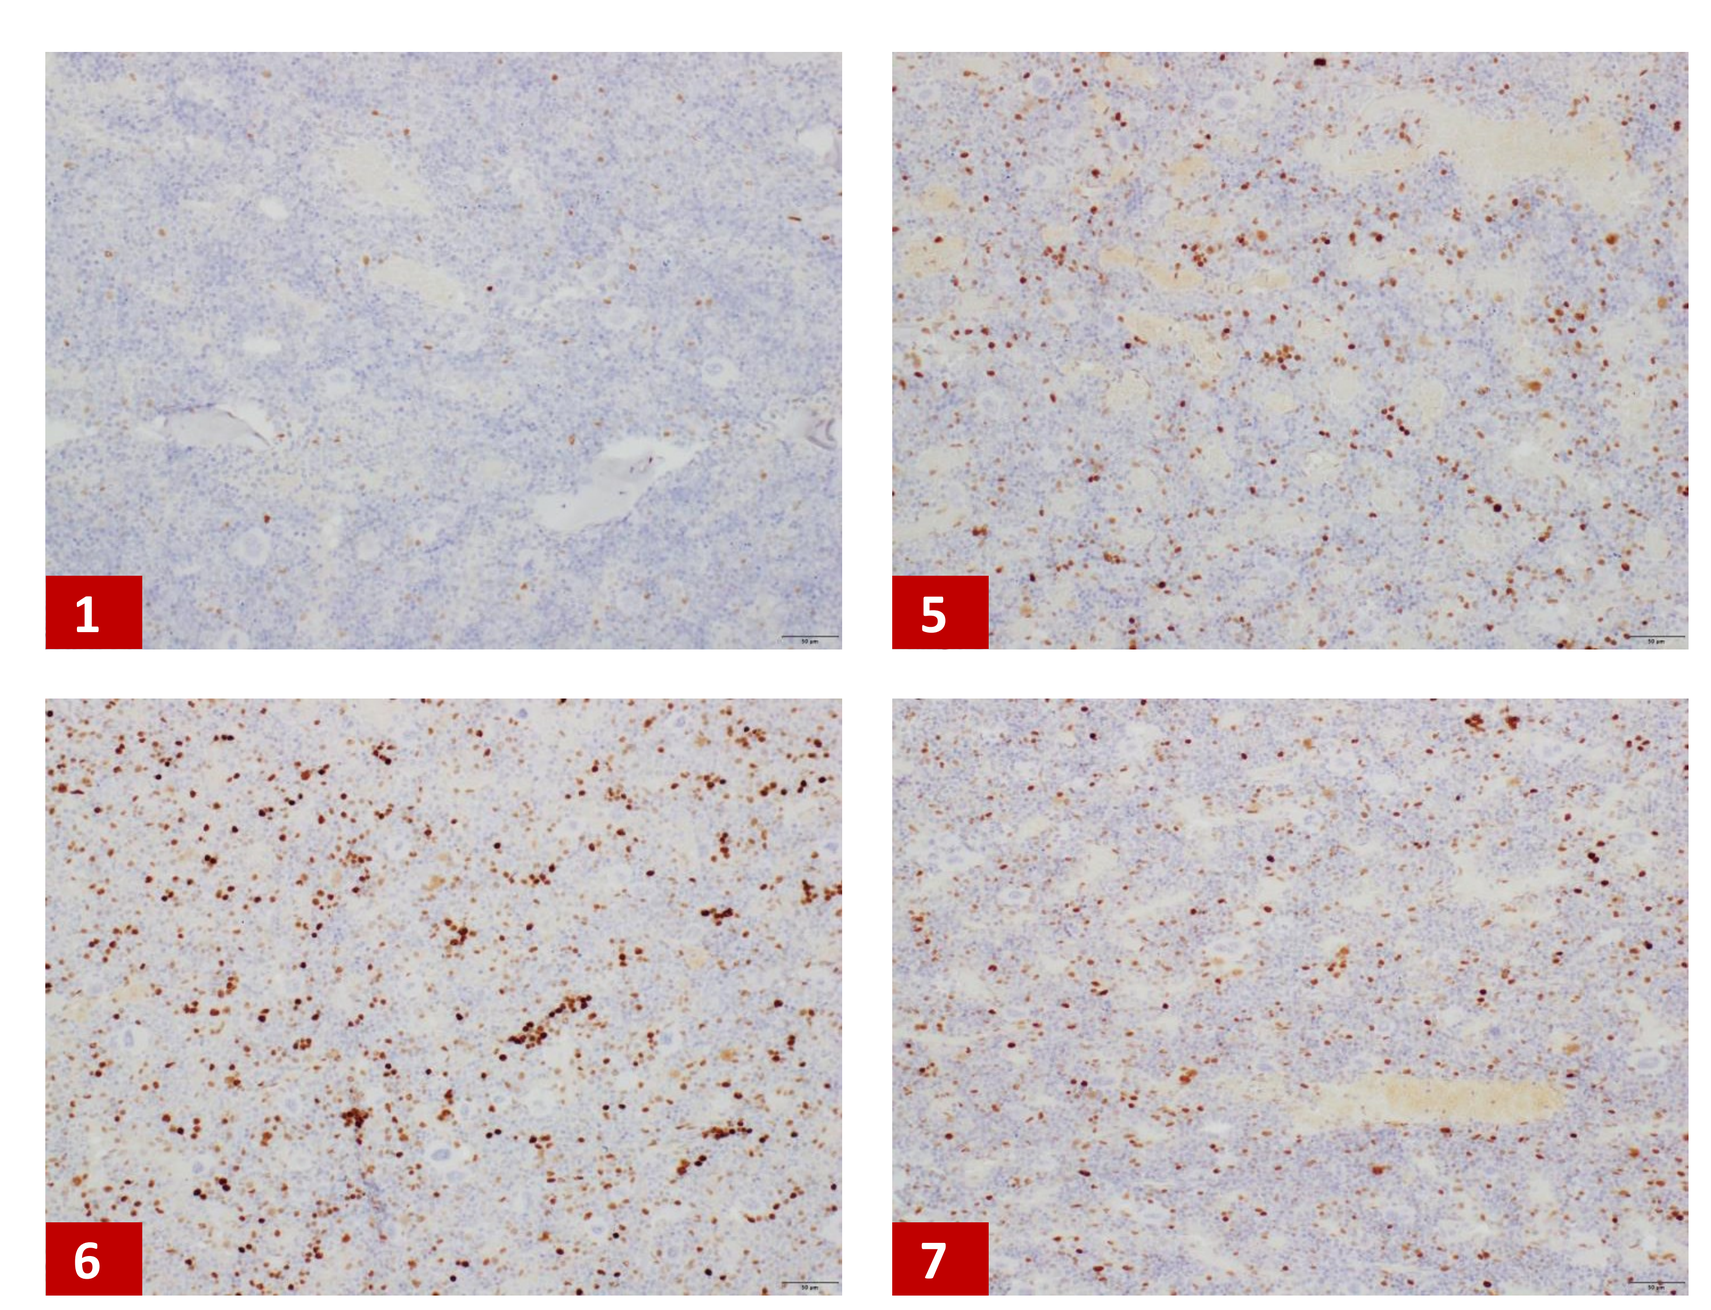
1
5
6
7

Dosing Relative to Sampling

Group	Treatment	-24	-16	-8	-1	0 hr
1	Vehicle					
2	ALRN-6924 2.4mg/kg					
3	ALRN-6924 2.4mg/kg					
4	ALRN-6924 2.4mg/kg					
5	ALRN-6924 2.4mg/kg					
6	ALRN-6924 2.4mg/kg					
7	ALRN-6924 2.4mg/kg					

Figure 7. Sparse Staining for cPARP in Bone Marrow with Treatment-dependent Trends for | -8 hr | and | -16, -8 hr | Schedules

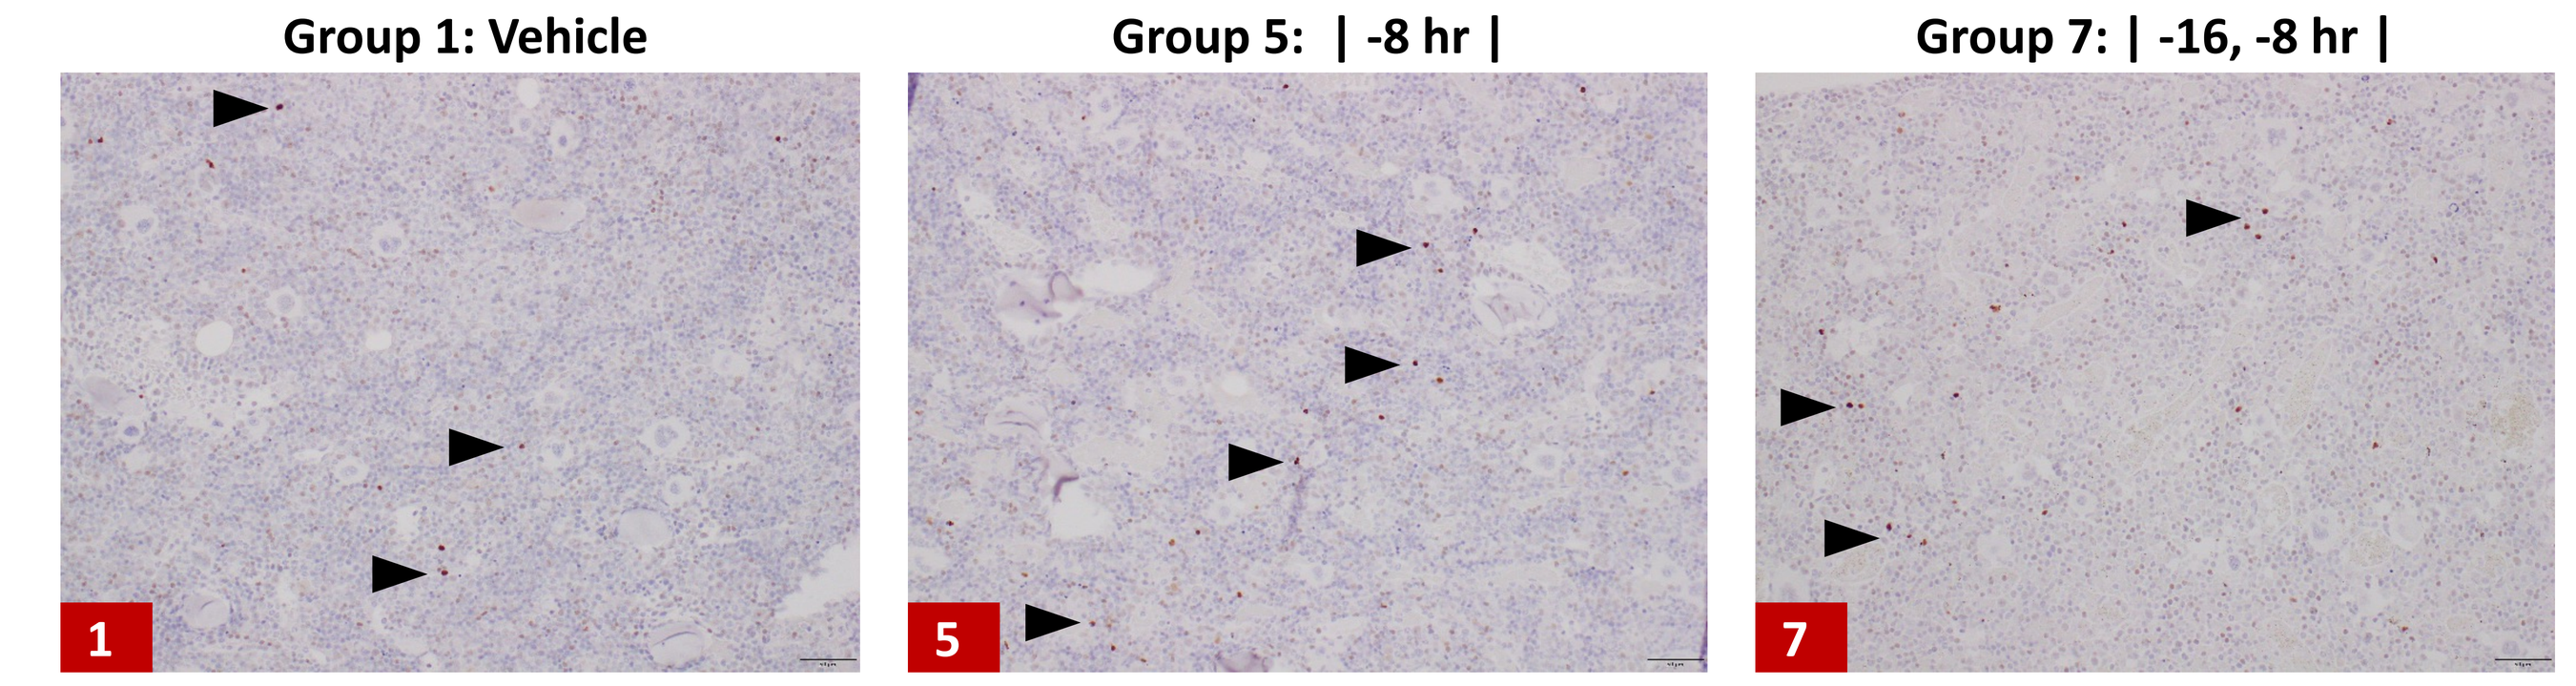
Group 1: Vehicle
Group 5: | -8 hr |
Group 7: | -16, -8 hr |
1
5
7

Figure 8. ALRN-6924 Upregulates p21, a Biomarker of p53-mediated Cell Cycle Arrest, in the GI Tract of Mice

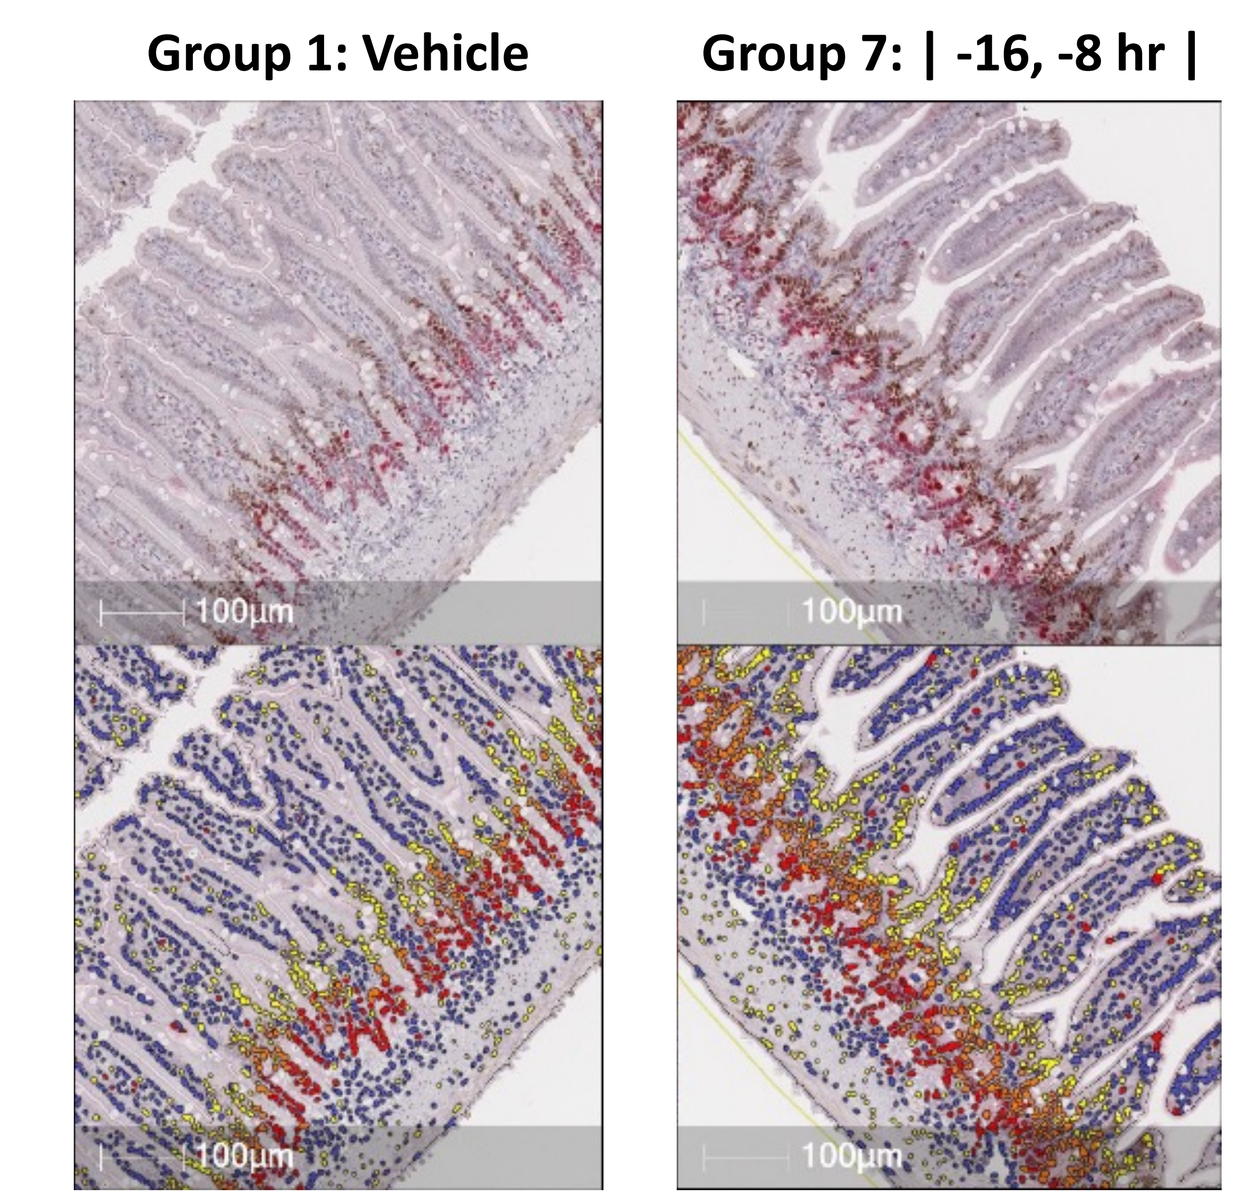
Group 1: Vehicle
Group 7: | -16, -8 hr |
100µm

Dual Chromogen Staining
Brown: p21-positive
Red: Ki67-positive

False Color Image
Blue: Negative nuclei
Yellow: p21-positive
Orange: p21 + Ki67
Red: Ki67-positive

Formalin-fixed jejunum were co-stained for p21 (cell cycle arrest) and Ki67 (proliferation) and analyzed by multicolor chromogenic IHC.

In vehicle-treated mice, Ki67 staining is marked in crypt cells, with mild p21 staining evident in rapidly cycling, transit-amplifying cells that differentiate as they migrate out of the crypts onto the villi.

Treatment with ALRN-6924 yields markedly more p21 staining in rapidly cycling, transit-amplifying cells than in placebo-treated mice.

Figure 9. Multiple ALRN-6924 Dose Regimens Were Evaluated for Reduction in Radiation-induced Mortality and BW Loss

Dosing Relative to Irradiation

Group	Treatment	-24	-16	-8	-1	0	8 hr
1	Irradiated Control						
2	Vehicle						
3	ALRN-6924 2.4mg/kg						
4	ALRN-6924 2.4mg/kg						
5	ALRN-6924 2.4mg/kg						
6	ALRN-6924 2.4mg/kg						
7	ALRN-6924 2.4mg/kg						
8	ALRN-6924 2.4mg/kg						
9	ALRN-6924 2.4mg/kg						

C57BL/6 mice (n=7/group) were exposed to a single dose of abdominally-targeted radiation at 15 Gy following one or more doses/schedules of ALRN-6924 and then monitored for body weight.

Figure 10. ALRN-6924 Protects Mice from Radiation-induced Toxicity, with | -8 hr |, | -24, -8 hr |, | -16, -8 hr | Schedules Best

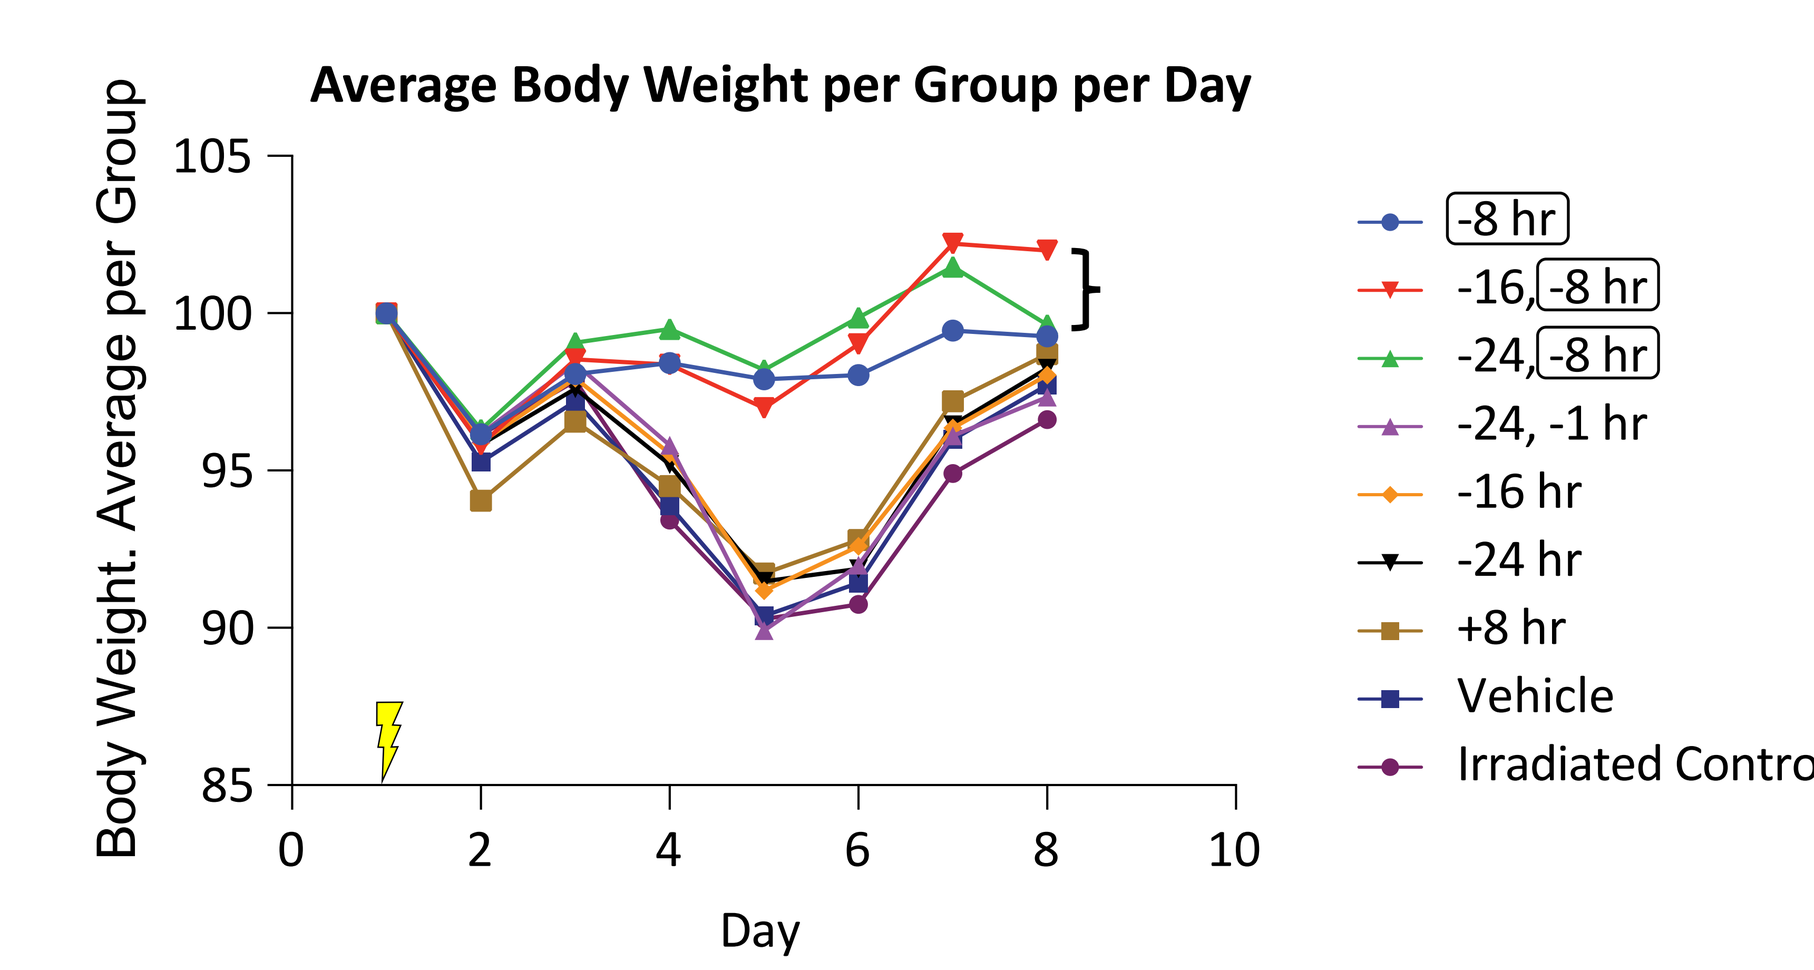
Average Body Weight per Group per Day
Body Weight. Average per Group
Day
-8 hr
-16, -8 hr
-24, -8 hr
-24, -1 hr
-16 hr
-24 hr
+8 hr
Vehicle
Irradiated Control

Body weight loss is significantly improved for mice receiving one or more ALRN-6924 doses at 8 hrs prior to irradiation

Conclusions

- ALRN-6924 mitigated toxicity in a mouse model of acute radiation injury.
- The observed radioprotection effect was correlated with cell cycle arrest in bone marrow after one or more doses of ALRN-6924.
- These results support further investigation of ALRN-6924 as a radioprotective agent.

63rd Annual Meeting of the American Society of Hematology (ASH) • December 11 - 14, 2021 • Atlanta, Georgia
Aileron Therapeutics, Inc. • 285 Summer Street, Boston, MA 02210 • Phone: 617-995-0900 | Fax: 617-995-2410 | Email: info@aileronrx.com • aileronrx.com