Harnessing the Anticancer Activity of the Stapled Peptide ALRN-6924, a Dual Inhibitor of MDMX and MDM2, Using Rational Combination Strategies for Breast Cancer and Other Malignancies

Allen Annis[1], Jian-Guo Ren[1], Luis A. Carvajal[1], Solimar Santiago[1], Narayana Narasimhan[1], David Sutton[1], Seyed Saeed Pairawan[2], Vincent Guerlavais[1], Funda Meric-Bernstam[2], Manuel Aivado[1]

[1]Aileron Therapeutics Inc., Watertown, MA, USA; [2]MD Anderson Cancer Center, Houston, TX, USA.

Background

The purpose was to identify rational anticancer drug combinations with ALRN-6924.

Material and Methods

ALRN-6924 is a cell-permeating, stabilized α-helical peptide that mimics the p53 tumor suppressor protein to disrupt its interactions with both its endogenous inhibitors, MDMX and MDM2. For p53 wild-type tumors, ALRN-6924 can restore p53-dependent tumor suppression leading to antitumor efficacy. ALRN-6924 was tested in combination with 29 drugs for synergistic *in vitro* anticancer activity. Select agents were further evaluated *in vivo*. Drugs listed in Table 1 were assayed in combination with ALRN-6924 using WST-1 and/or CyQUANT cell viability assays in immortalized TP53 wild-type cancer cell lines. Synergy was quantified by the Chou-Talalay combination index (CI) method. *In vivo* ALRN-6924 combinations with palbociclib, abemaciclib, and nab-paclitaxel were tested in the MCF-7 breast cancer model in athymic nude mice.

Results

All the drugs evaluated except dexamethasone were additive or synergistic with ALRN-6924 *in vitro*; no antagonism was observed. Pharmacodynamic biomarkers indicate on-mechanism activity. In MCF-7 xenografts tumor growth inhibition was improved when ALRN-6924 was given in combination with palbociclib, abemaciclib, or nab-paclitaxel compared to single agent therapy. Body weights and mortality data suggest ALRN-6924 and combinations with nab-paclitaxel and palbociclib were tolerated at the doses tested. The combination with abemaciclib was tolerated with interruption and dose-reduction.

Figure 1: The Cell-Permeating Stabilized α-helical Peptide, ALRN-6924, is a First-in-Class Dual Inhibitor of MDMX and MDM2

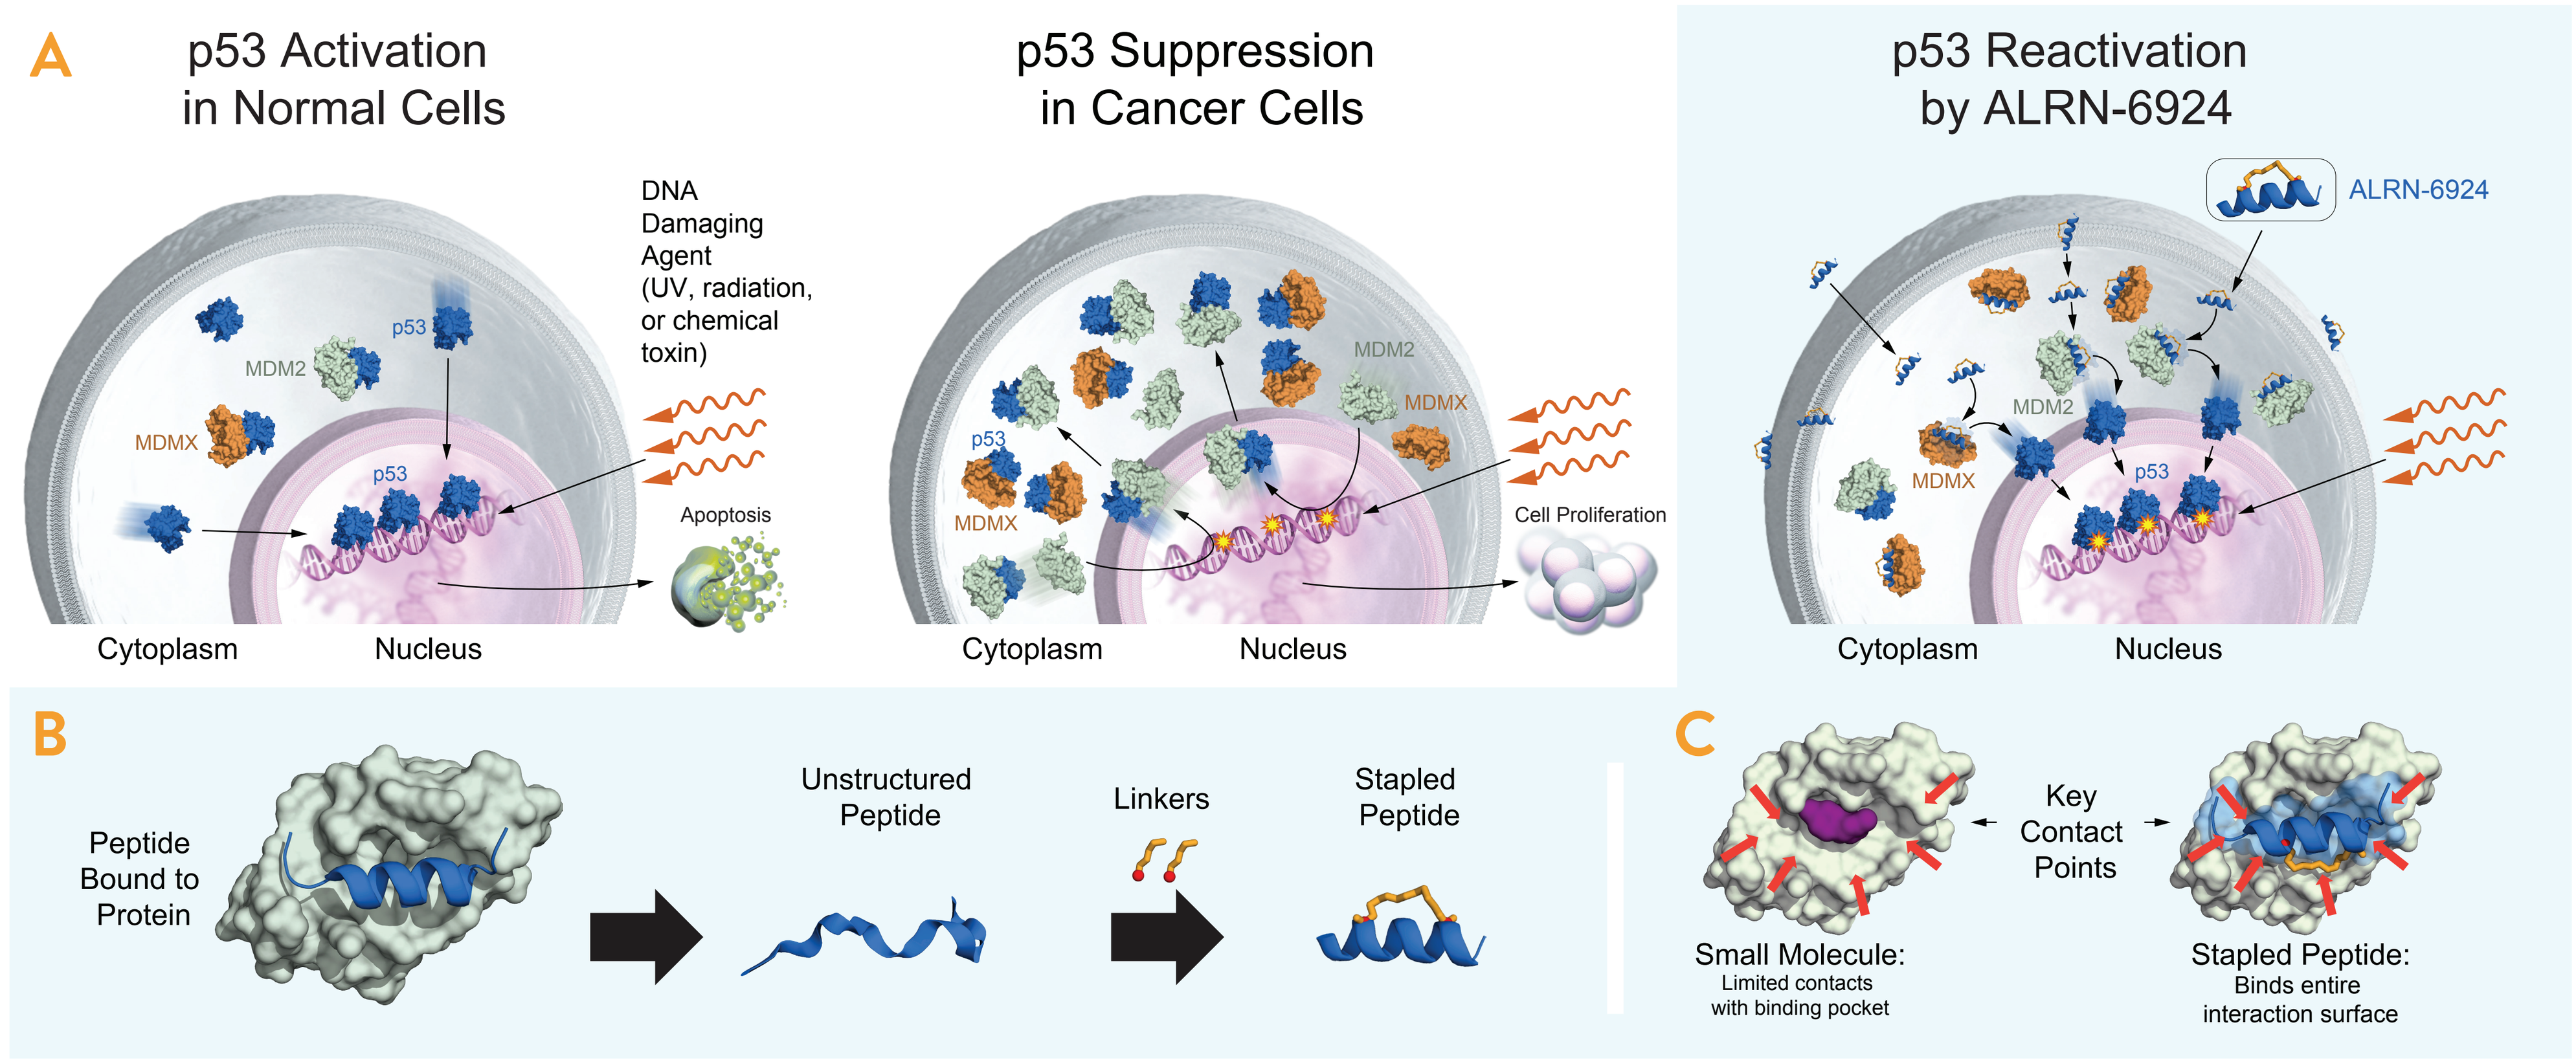
A
p53 Activation in Normal Cells
p53 Suppression in Cancer Cells
p53 Reactivation by ALRN-6924
ALRN-6924
DNA Damaging Agent (UV, radiation, or chemical toxin)
MDM2
MDMX
p53
Apoptosis
Cell Proliferation
Cytoplasm
Nucleus
B
Peptide Bound to Protein
Unstructured Peptide
Linkers
Stapled Peptide
C
Key Contact Points
Small Molecule: Limited contacts with binding pocket
Stapled Peptide: Binds entire interaction surface

A) The tumor suppressor p53 is one of the most pursued targets in oncology, playing a central role inducing cell cycle arrest, apoptosis, senescence, autophagy, cellular metabolism and immune surveillance in response to cellular stresses such as DNA damage and oncogenic signals[1]. B) ALRN-6924, a cell-permeating, stapled α-helical peptide that has demonstrated anticancer activity as monotherapy in clinical trials, mimics the p53 tumor suppressor protein to disrupt its interactions with both its endogenous inhibitors, MDMX and MDM2[2,3]. Stapled peptides mimic natural peptide sequences at the interface of protein-protein interactions, displaying a larger surface area of interaction with its target, and providing superior binding properties which reduce off-target effects and the risk of acquiring mutations associated with resistance. C) Furthermore, like natural protein sequences, a peptide can engage with ≥2 targets, e.g. MDMX + MDM2.

Table 1: ALRN-6924 Synergizes with Commonly Used Pathway-Selective and Chemotherapy Agents

Combination indices for the indicated cell lines and corresponding test article are shown. Cells were dosed with varying concentrations of ALRN-6924, the indicated test article alone, or in combination with ALRN-6924. Cell viability was measured using a standard colorimetric MTT assay. Drug combination index (CI) analysis was conducted following the Chou-Talalay method using CompuSyn. Synergism and antagonism were described based on the CI values according to the Chou and Talalay guidelines.

* *In vitro* CI value at IC_{75} or ‡IC_{50}, average ± SD of ≥ 2 experiments. CI values: 0–0.1, very strong synergism; 0.1–0.3, strong synergism; 0.3–0.7, synergism; 0.7–0.85, moderate synergism; 0.85–0.90, slight synergism; 0.90–1.10, nearly additive; 1.10–1.20, slight antagonism; 1.20–1.45, moderate antagonism; 1.45–3.3, antagonism; 3.3–10, strong antagonism; 10, very strong antagonism). †Single experiment.

Cancer	Cell line	Drug	CI*
Breast Cancer	MCF-7	palbociclib	0.45 ± 0.17
		abemaciclib	0.57 ± 0.23
		ribociclib	0.39 ± 0.08
		paclitaxel	0.80 ± 0.05
		docetaxel	0.37 ± 0.02
		eribulin	0.66 ± 0.36
		everolimus	0.59 ± 0.28
		fulvestrant	0.41 ± 0.17‡
		carboplatin	0.49 ± 0.04
		gemcitabine	0.58 ± 0.05
		dexamethasone	No change
Colon Cancer	HCT-116	5-FU	0.07 ± 0.03
Melanoma	A375	trametinib	0.41 ± 0.12
		dabrafenib	0.33 ± 0.15
		vemurafenib	0.48 ± 0.20
	MEL-JUSO	binimetinib	0.32 ± 0.10
		pimasertib	0.39 ± 0.08
		selumetinib	0.49 ± 0.08
T Cell Lymphoma	MOLT-3	romidepsin	0.38 ± 0.00
B Cell Lymphoma	DOHH-2	vincristine	0.09 ± 0.06
		cyclophosphamide	0.11 ± 0.13
		rituximab	0.40†
		doxorubicin	0.04†
	JVM2	ibrutinib	0.59 ± 0.27
Acute Leukemia	MV-4-11	cytarabine/Ara-C	0.68 ±0.02
		azacitidine	0.49 ± 0.32
		decitabine	1.08 ± 0.07
		midostaurin	0.83 ± 0.04
		venetoclax	0.20 ± 0.03

Figure 2: ALRN-6924 Displays Additive to Synergistic Antitumor Activity in Combination with Paclitaxel *in vitro*

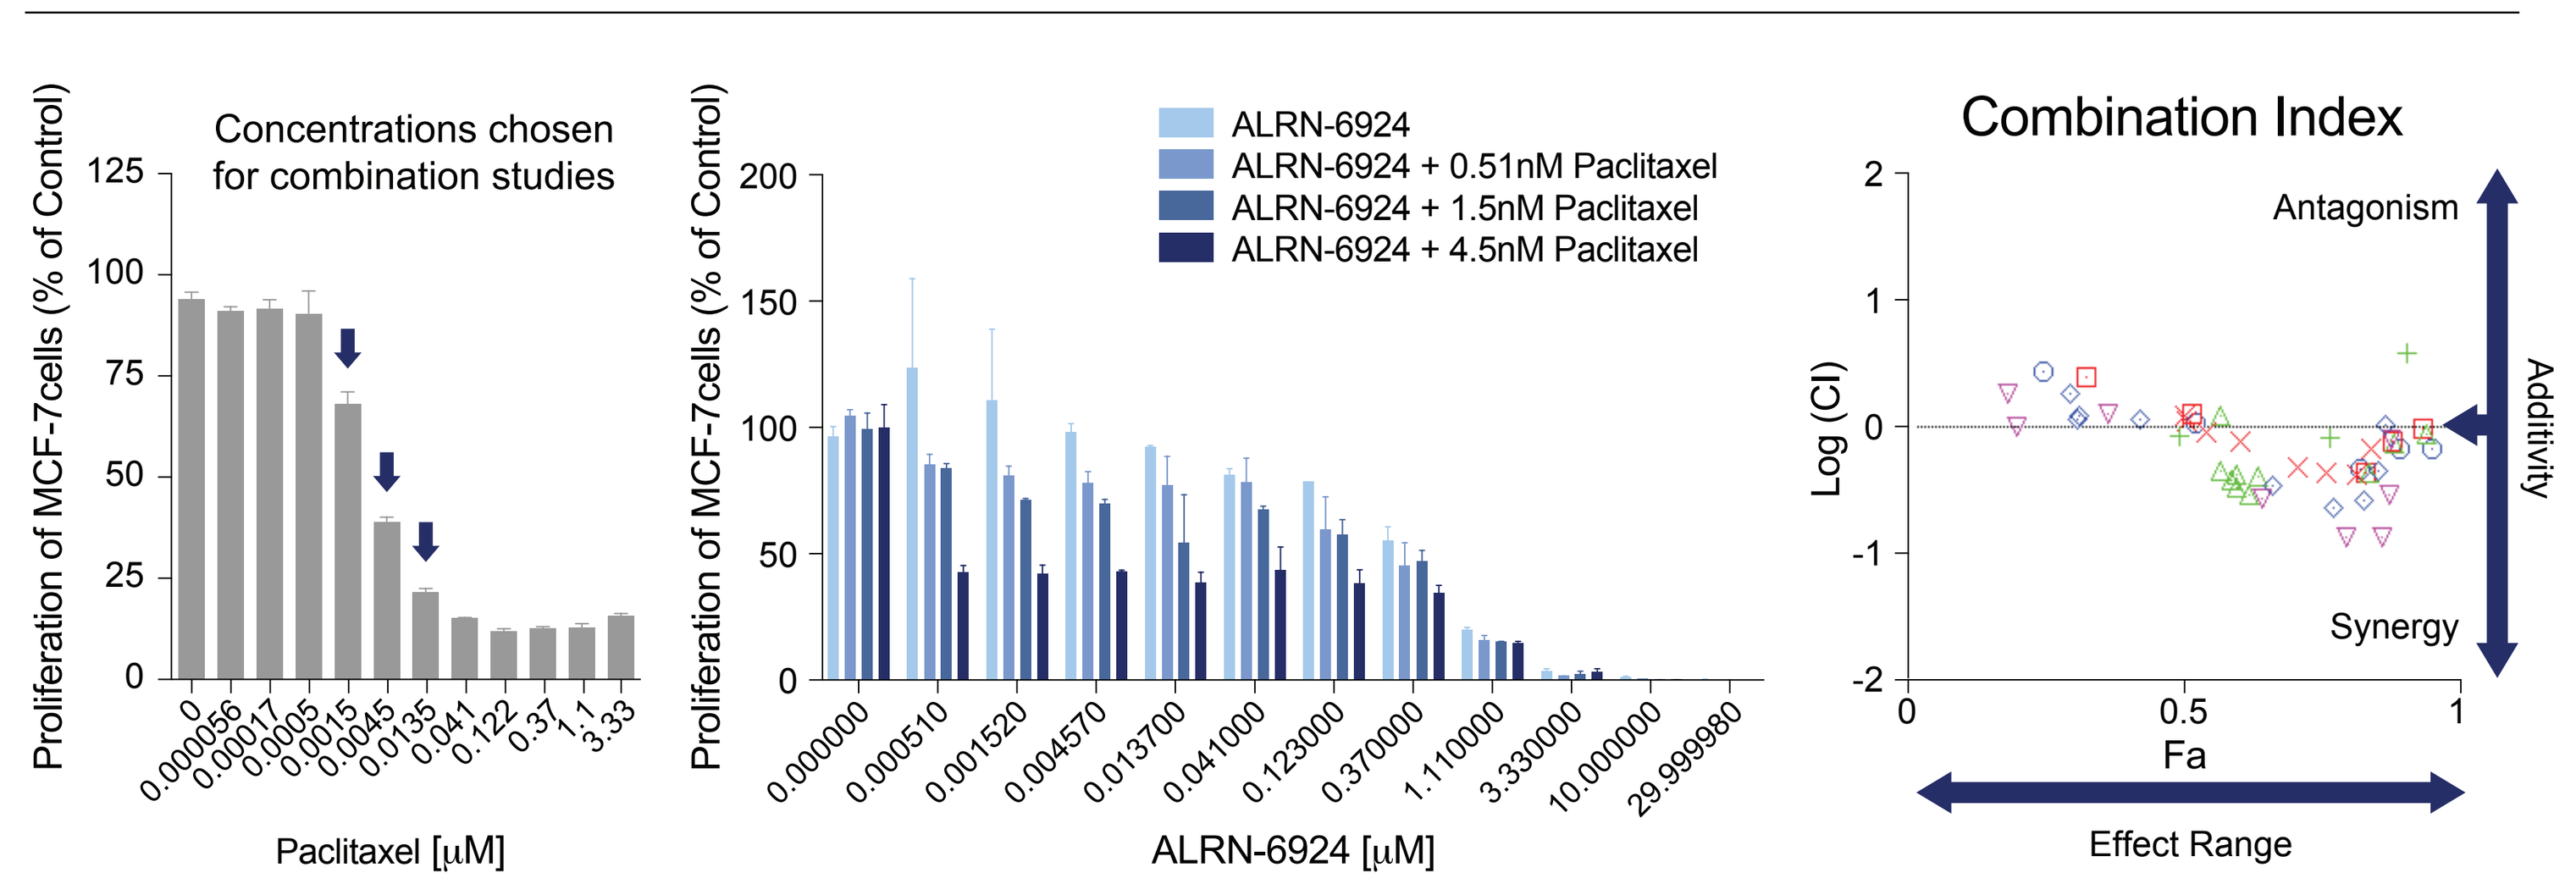
A
B
MCF-7 Cells
C
Concentrations chosen for combination studies
Proliferation of MCF-7cells (% of Control)
Paclitaxel [μM]
ALRN-6924
ALRN-6924 + 0.51nM Paclitaxel
ALRN-6924 + 1.5nM Paclitaxel
ALRN-6924 + 4.5nM Paclitaxel
ALRN-6924 [μM]
Combination Index
Antagonism
Additivity
Synergy
Log (CI)
Fa
Effect Range

A) Cell viability in response to varying concentrations of paclitaxel (blue arrows denote concentrations chosen for combination studies). B) Proliferation of MCF-7 cells treated with the indicated dose of paclitaxel and varying concentrations of ALRN-6924. C) Combination indices for the drug combination of ALRN-6924 and paclitaxel were determined as described in Table 1.

Figure 3: ALRN-6924 Enhances the Antitumor Activity of Nab-Paclitaxel in the MCF-7 Breast Cancer Xenograft Model

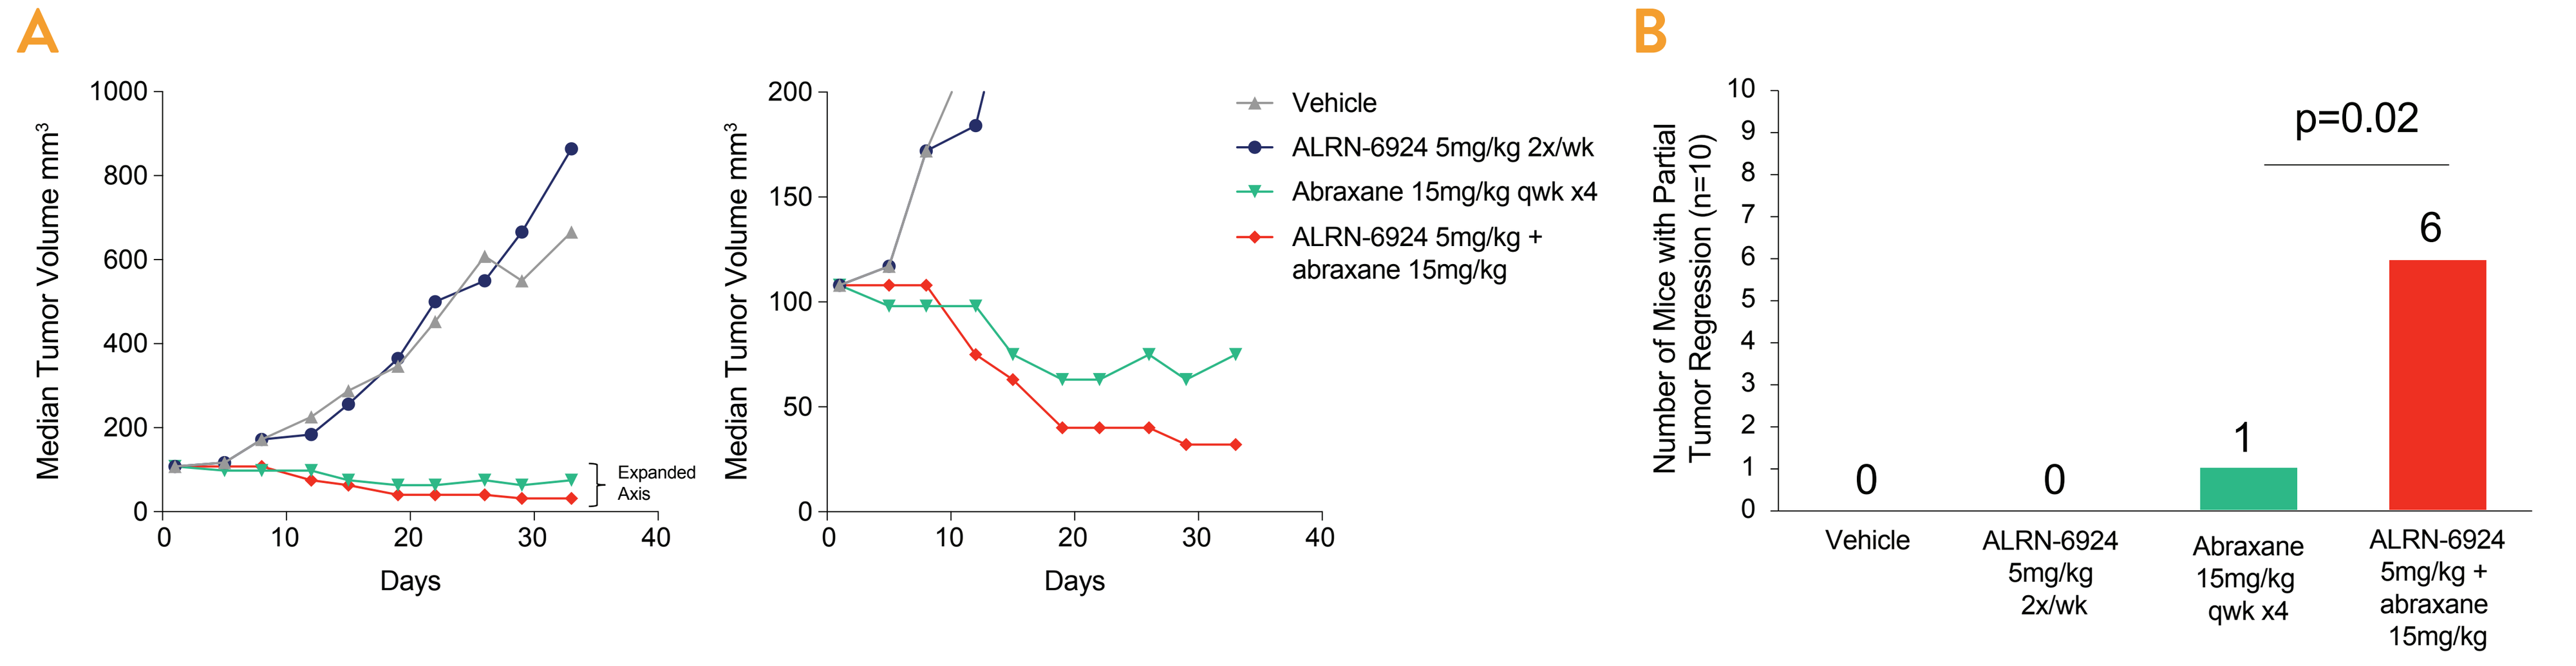
A
Median Tumor Volume mm3
Days
Expanded Axis
Vehicle
ALRN-6924 5mg/kg 2x/wk
Abraxane 15mg/kg qwk x4
ALRN-6924 5mg/kg + abraxane 15mg/kg
B
Number of Mice with Partial Tumor Regression (n=10)
p=0.02
Vehicle
ALRN-6924 5mg/kg 2x/wk
Abraxane 15mg/kg qwk x4
ALRN-6924 5mg/kg + abraxane 15mg/kg

A) Athymic nude mice with established tumors (n = 10 per group) were treated for 4 weeks with ALRN-6924 twice-weekly alone or in combination with weekly doses of nab-paclitaxel. Compounds were co-administered intravenously at the indicated doses. B) Objective tumor responses on d32 (partial regression = 3 consecutive measurements <50% of starting volume). Studies were approved by the Institutional Animal Care and Use committee at Charles River Laboratories, Morrisville, N.C

Figure 4: ALRN-6924 Displays Additive to Synergistic Antitumor Activity in Combination with Palbociclib *in vitro*

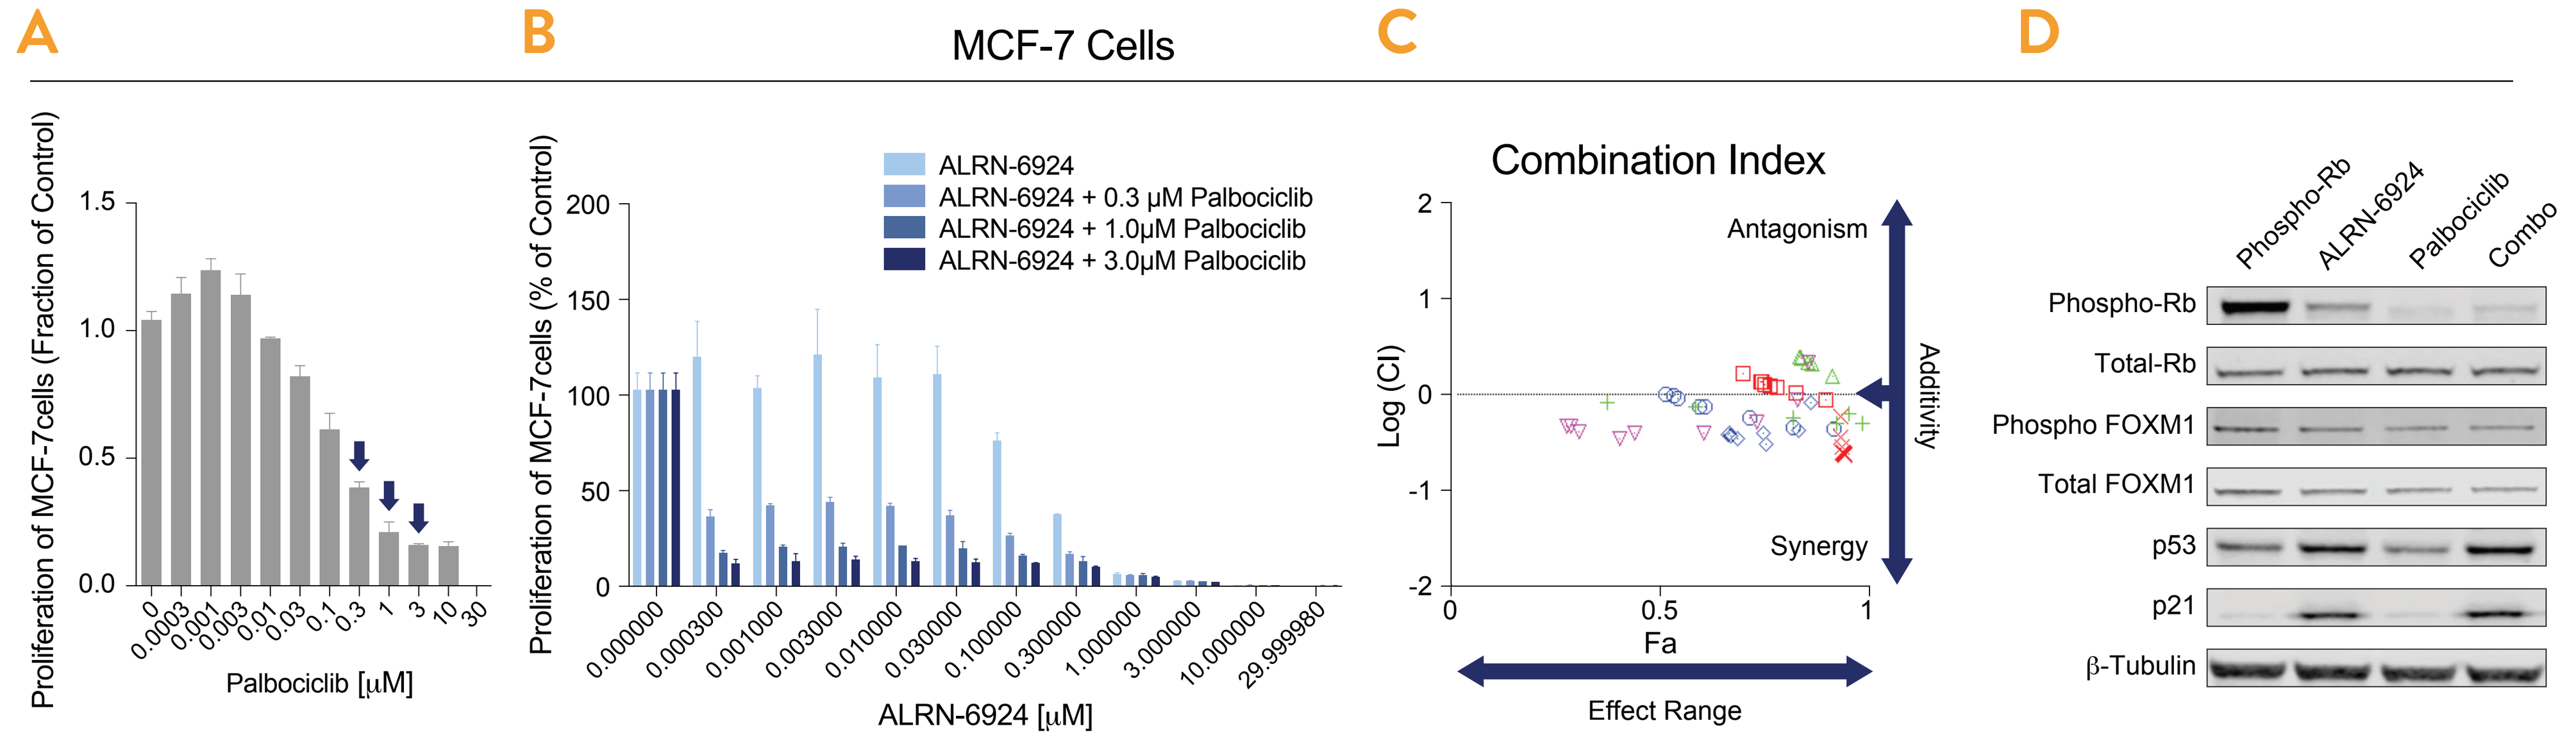
A
B
MCF-7 Cells
C
D
Proliferation of MCF-7cells (Fraction of Control)
Palbociclib [μM]
ALRN-6924
ALRN-6924 + 0.3 μM Palbociclib
ALRN-6924 + 1.0μM Palbociclib
ALRN-6924 + 3.0μM Palbociclib
Proliferation of MCF-7cells (% of Control)
ALRN-6924 [μM]
Combination Index
Antagonism
Additivity
Synergy
Log (CI)
Fa
Effect Range
Phospho-Rb
ALRN-6924
Palbociclib
Combo
Phospho-Rb
Total-Rb
Phospho FOXM1
Total FOXM1
p53
p21
β-Tubulin

A) Cell viability in response to varying concentrations of palbociclib (blue arrows denote concentrations chosen for combination studies). B) Proliferation of MCF-7 cells treated with the indicated dose of palbociclib and varying concentrations of ALRN-6924. C) Combination indices for the drug combination of ALRN-6924 and palbociclib were determined as described in Table 1. D) Protein expression of pharmacodynamic biomarkers demonstrating on-mechanism activity of test articles as single agents and in combination.

Figure 5: ALRN-6924 Increases Progression Free Survival in Combination with Palbociclib and Abemaciclib in the MCF-7 Breast Cancer Xenograft Model Compared to Single Agents

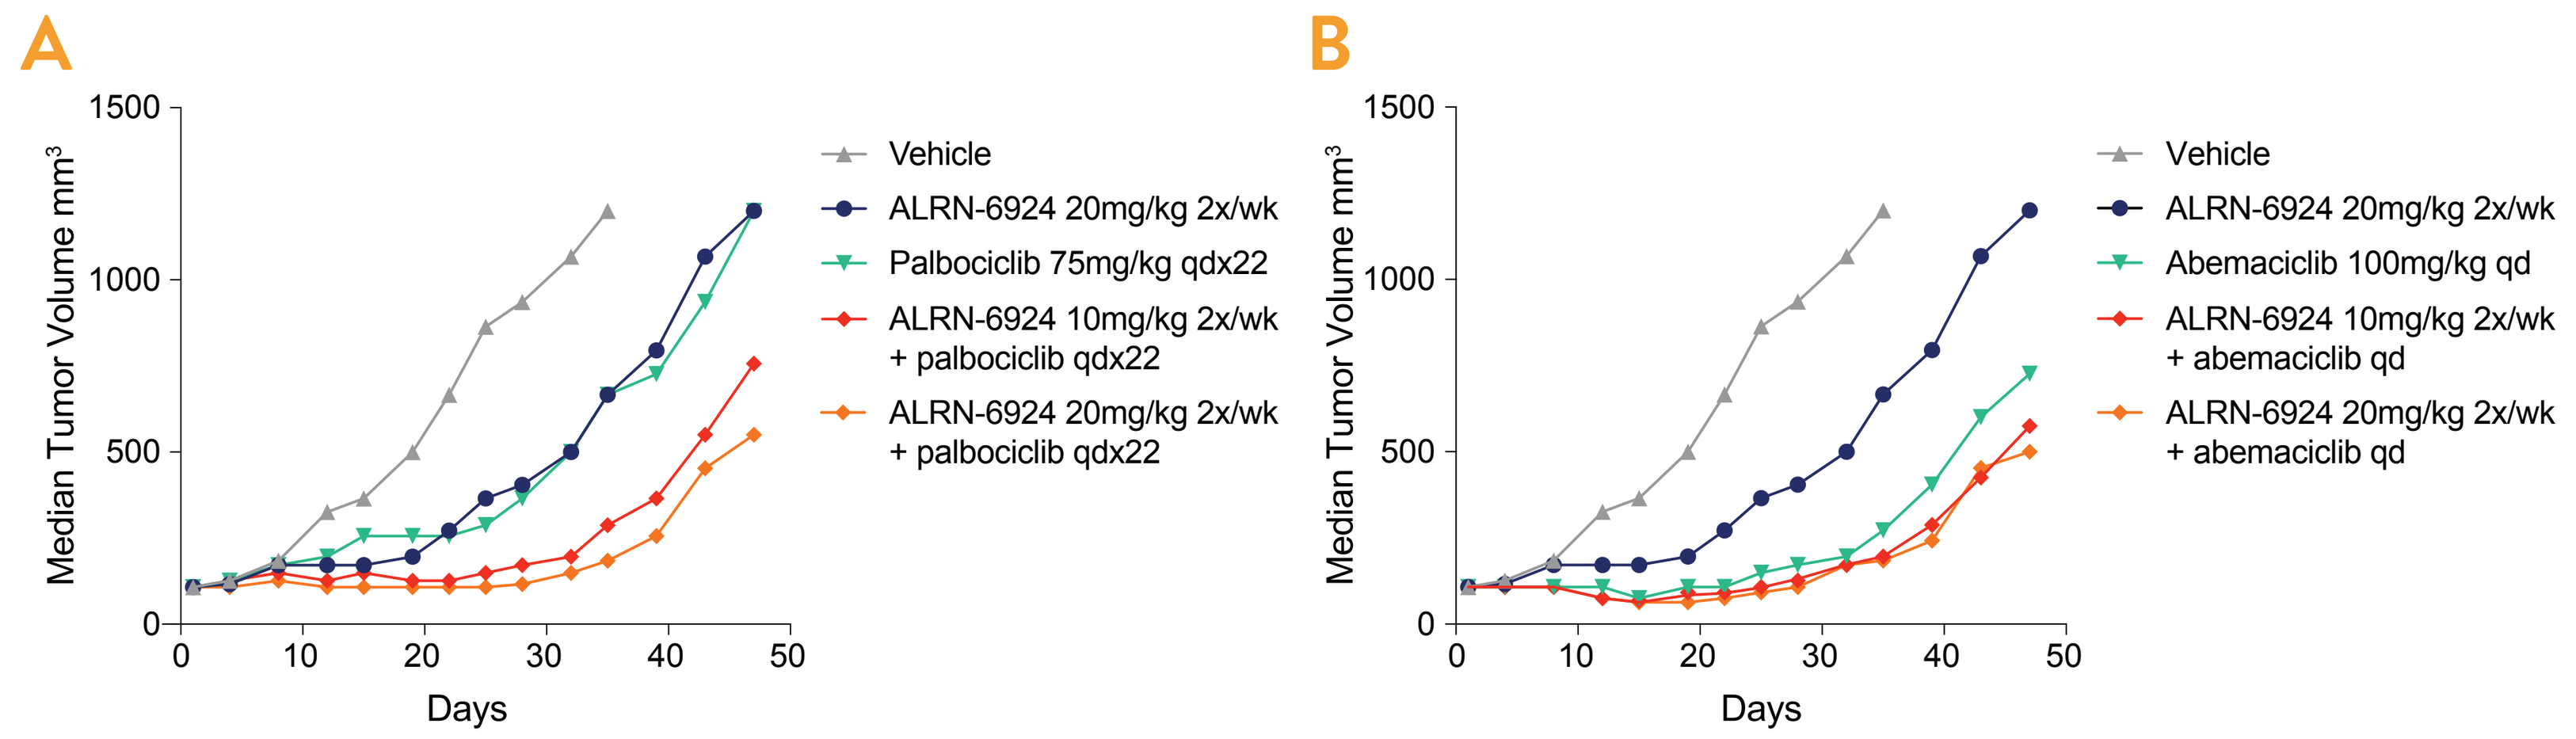
A
Median Tumor Volume mm3
Days
Vehicle
ALRN-6924 20mg/kg 2x/wk
Palbociclib 75mg/kg qdx22
ALRN-6924 10mg/kg 2x/wk + palbociclib qdx22
ALRN-6924 20mg/kg 2x/wk + palbociclib qdx22
B
Vehicle
ALRN-6924 20mg/kg 2x/wk
Abemaciclib 100mg/kg qd
ALRN-6924 10mg/kg 2x/wk + abemaciclib qd
ALRN-6924 20mg/kg 2x/wk + abemaciclib qd

A) Athymic nude mice with established tumors were treated with ALRN-6924 alone (intravenously twice weekly for 3 weeks) or in combination with palbociclib (orally daily for 22 days) or B) abemaciclib (orally daily - 100mg/kg. Dosing was stopped on day 14 due to weight loss. After a brief treatment holiday, dosing resumed on day 18 at a lower dose – 75mg/kg for 5 days.) Studies were approved by the Institutional Animal Care and Use committee at Charles River Laboratories, Morrisville, N.C.

Conclusions

ALRN-6924 is a first-in-class clinical stage MDMX/2 dual inhibitor displaying promising anticancer activity in patients[2,3]. Recently, ALRN-6924 was shown to display strong on-mechanism anti-tumor activity in preclinical models of AML[4] and PTCL[5]. In the current study, we demonstrate that ALRN-6924 can be rationally combined with pathway-selective and chemotherapy agents. These results, plus promising ALRN-6924 safety and antitumor activity as a monotherapy[3], support the development of combination regimens for breast cancer and other malignancies.

References

1. Bieging KT et al., Unravelling mechanisms of p53-mediated tumour suppression, *Nat Rev Cancer.* 2014; 14 (5):359-70
2. Meric-Bernstam F. et al., Phase I trial of a novel stapled peptide ALRN-6924 disrupting MDMX and MDM2-mediated inhibition of WT p53 in patients with solid tumors and lymphomas, *J Clin Oncol.* 2017; 35 (15):2505-2505
3. Meric-Bernstam F. et al., ASCO 2017 slides presentation: http://share.aileronrx.com/presentations/ALRN-6924_Final_Slides_ASCO_02Jun2017.pdf
4. Carvajal L.A. et al. Dual inhibition of MDMX and MDM2 as a therapeutic strategy in leukemia, *Sci Transl Med.* 2018; 10 (436)
5. Ng SY, et al., Targetable vulnerabilities in T- and NK-cell lymphomas identified through preclinical models. *Nat Commun.* 2018; 9 (1): 2024

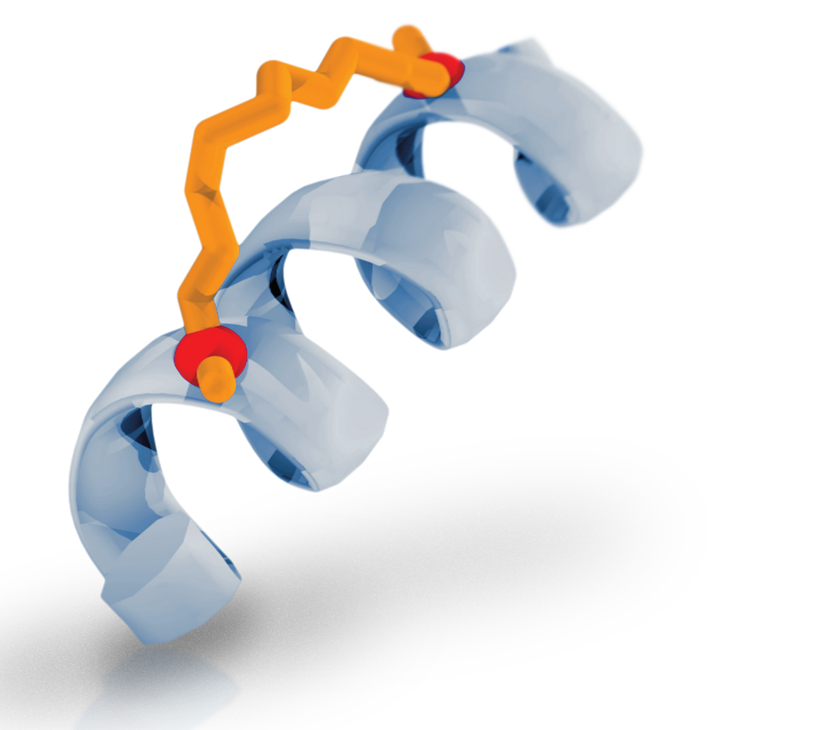

AILERON THERAPEUTICS 490 Arsenal Way - Watertown, MA 02472 USA | Phone: 617-995-0900 | Fax: 617-995-2410 | info@aileronrx.com

2018 EORTC-NCI-AACR Molecular Targets and Cancer Therapeutics Symposium